Evaluation of Tandem Dual Stationary Phases for Mass-Directed SFC Purification of Small Molecule Compound Libraries

Vivi Lazarescu, Mark J. Mulvihill and Lifu Ma

Department of Cancer Chemistry, OSI Pharmaceuticals, Farmingdale, NY 11735

Abstract

Recently, we reported the utility of the 4-ethylpyridine column as a preferred SFC stationary phase for the purification of small molecule compound libraries in our discovery laboratories. In our quest to identify a more versatile stationary phase system with improved chromatographic resolution and selectivity power, we investigated a dual column approach which pairs a 4-ethylpyridine stationary phase with a selectivity-orthogonal achiral or chiral stationary phase. The combinative effect of orthogonal stationary phases on chromatographic performance and library separation will be discussed.

Background

With the goal of shortening purification cycle times and increasing library output, we recently developed a "Universal One-Column-For-All" SFC approach[1], which mimics the common C-18 practice in reversed phase HPLC by adopting a single semi-preparative stationary phase for the purification of internal library compounds (Scheme 1). Among eight stationary phases tested, 4-Ethylpyridine (4-EP) emerged as the preferred stationary phase, affording the highest % resolution with a ~95% success rate. In short, 77% of compounds were resolved and 17% were partially resolved in the analytical screen. Further resolution could be achieved in the preparative stage with the combinative use of compound-specific gradients methods and Boolean Logical fractionation (Fig. 1).

Scheme 1. "Universal One-Column-For-All" SFC Approach

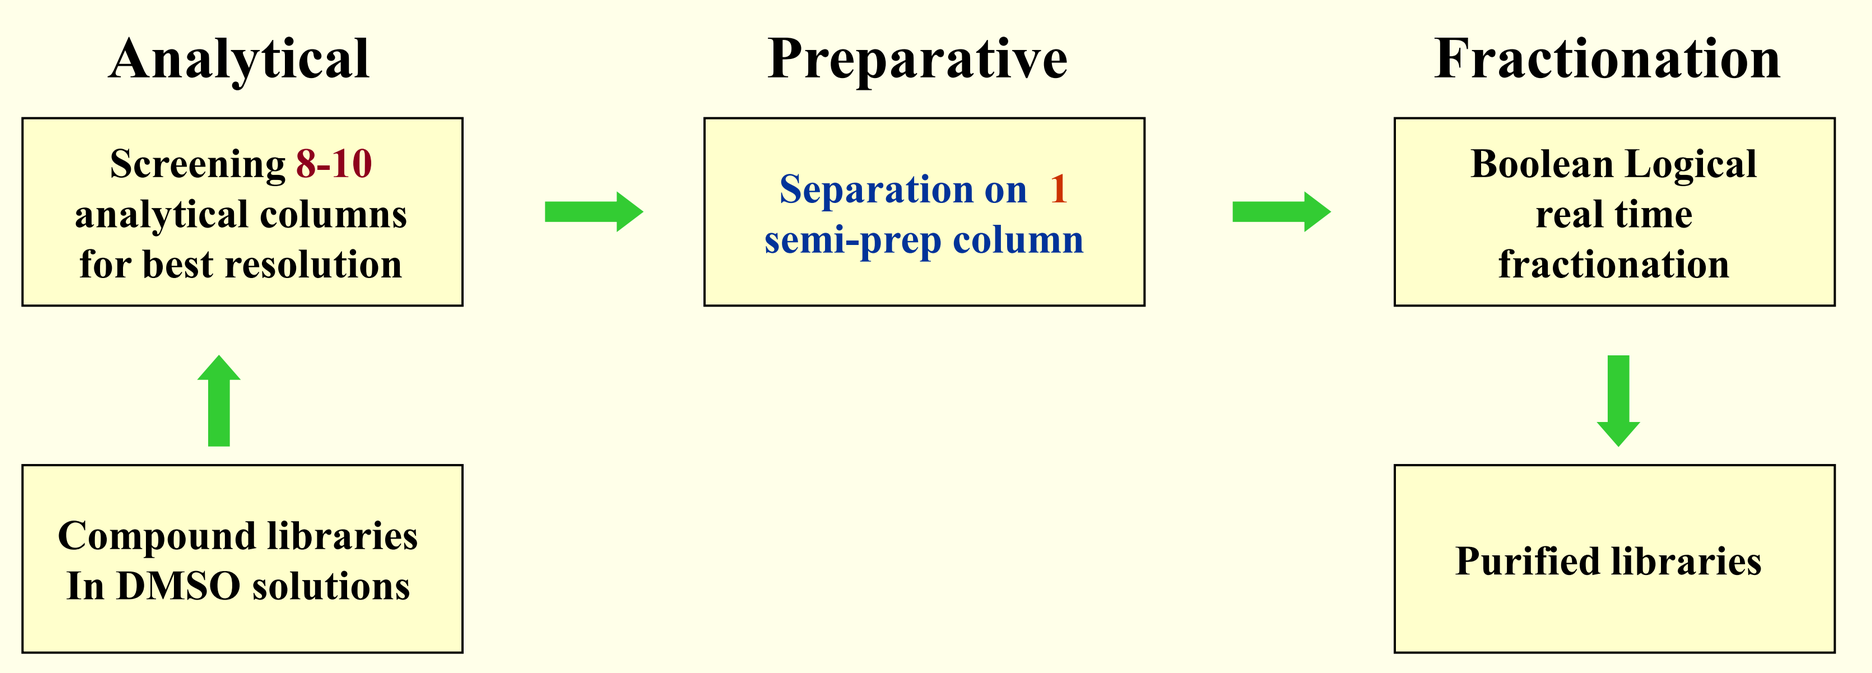
Analytical
Preparative
Fractionation
Screening 8-10 analytical columns for best resolution
Separation on 1 semi-prep column
Boolean Logical real time fractionation
Compound libraries In DMSO solutions
Purified libraries

- A set of analytical stationary phases are bundled together to identify the best one
- Rank order the columns based on statistical analysis of the chromatographic data
- A generic analytical gradient method ensures the targets & impurities all eluted out
- Tune additive composition to maximize column selectivity and resolution
- One semi-preparative stationary phase is used for all library compounds

Figure 1. SFC Column Performance Profile

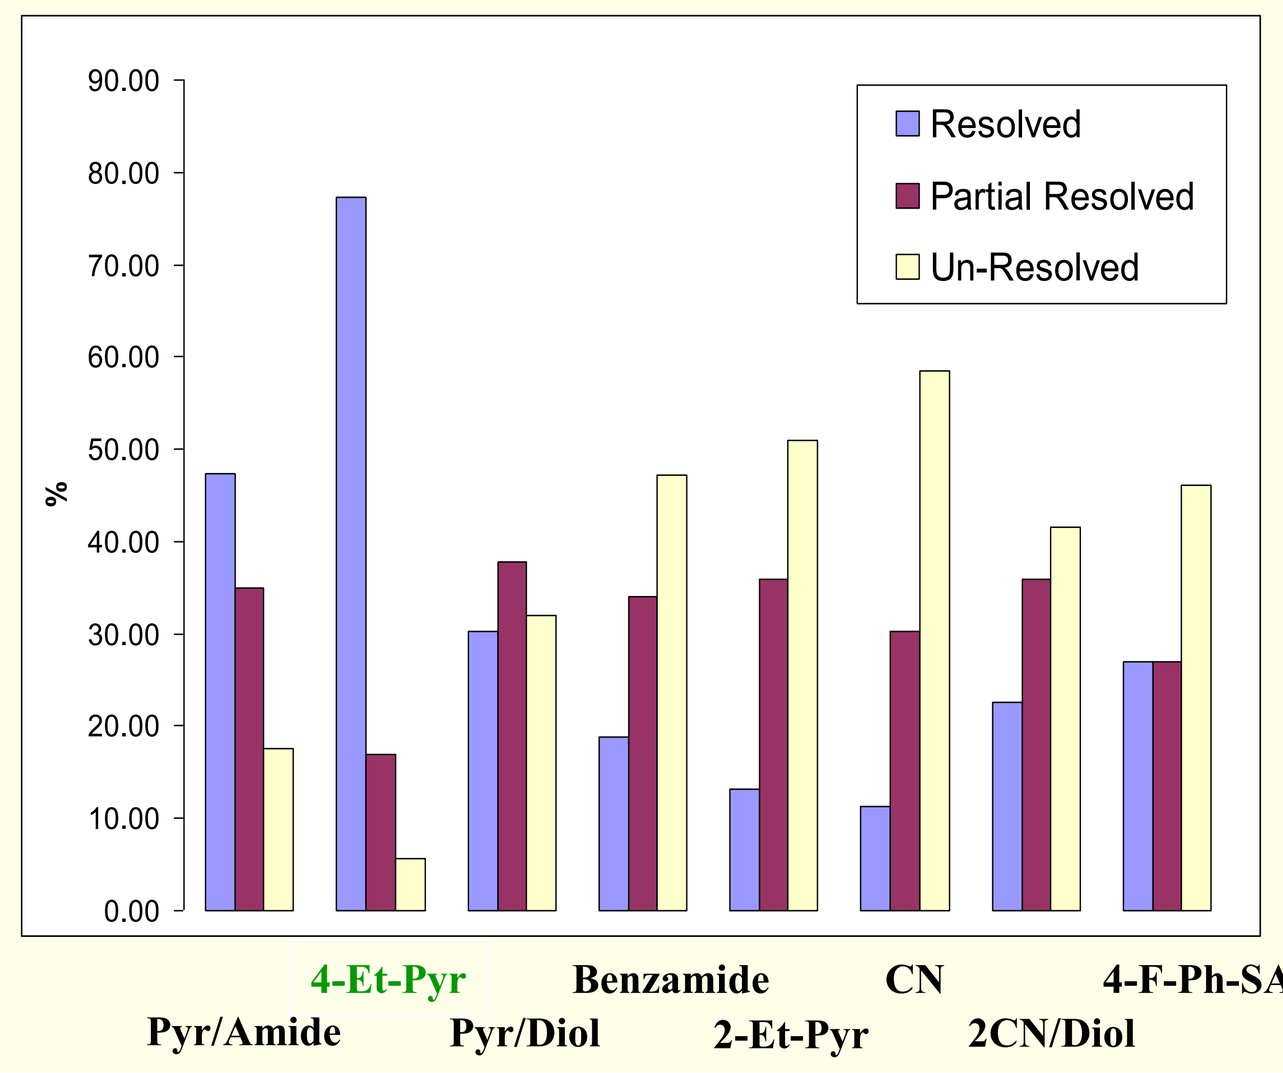
Resolved
Partial Resolved
Un-Resolved
%
4-Et-Pyr
Benzamide
CN
4-F-Ph-SAM
Pyr/Amide
Pyr/Diol
2-Et-Pyr
2CN/Diol

Following up the one-column approach with the aim to maximize both robustness and success rate applicable to all library mixtures and specifically "unresolvable mixtures", we report our feasibility evaluation of a dual tandem column approach which couples a 4-EP SFC column with a selectivity-orthogonal stationary phase.[1] The rationale behind this approach stems from our empirical chromatographic experience that "dissimilar stationary phases" such as Benzamide, 4-Fluorophenylsulfonamide (4-F-Ph-SAM), and 2CN/Diol are somewhat "orthogonal" to the basic 4-EP column in chromatographic selectivity and resolution power, presumably due to their complementary non-basic stationary phase-retention mechanism. A chiral stationary phase, CHIRALPACK IA was also included as we discovered it to be an appropriate stationary phase for achiral purification of challenging mixtures such as those containing structural isomers.[2,3] Similar coupling of achiral/chiral stationary phases for stereoisomeric separation has been reported.[4] The number of theoretical plates can be increased by sequentially coupling two columns while avoiding pressure drop limitations (due to the lower viscosities of SFC fluids) and reducing the likelihood of co-elution of products and impurities. For chromatographic evaluation, only scalable performances have been assessed therefore maintaining maximum resolution of impurities from the desired target and preserving peak shape.

Experimental

Instrumentation

Analytical SFC-MS system: Waters/Thar Technologies (Pittsburg, Pennsylvania), equipped with a Waters ZQ mass spectrometer.

Experimental Conditions

- **The crude samples from internal compound libraries were screened on four sets of stationary phases using standardized SFC conditions as described in Table 1.**
- **The achiral SFC analytical columns, 4-EP, Benzamide, 4-F-Ph-SAM, 2-CN/DIOL, 5 µm, 4.6 × 150 mm, were purchased from Princeton Chromatography Inc, while the CHIRALPACK IA, 5 µm, 4.6 × 100 mm, SFC chiral column was purchased from Chiral Technology, Inc.**
- **The analytical screening was performed under gradient conditions with 0.2% additive of IPAmine in methanol as modifier. In our hands, the use of 0.2% IPAmine helps achieve improved peak shape and separation of basic and/or halogenated mixtures, providing both good ionization patterns and enhanced loading capacity.**

Table 1. Experimental Conditions

Columns Tested	4-EP; Benzamide; 4-F-Ph-SAM; 2CN/Diol; IA
Mobile Phase	CO_2 / MeOH with 0.2% IPAmine
Temperature	40°C
Gradient	10 to 60% MeOH with 0.2% IPAmine in 5 min
Flow Rate	4 mL/min
Injection Volume	10 µL of ~200 mg/mL in DMSO

Results and Discussion

- **The initial chromatographic parameters chosen have been reported previously[1].**
- **Figures 2, 3 and 4 illustrate the typical chromatograms of library compounds on a 4-EP single column *versus* four achiral/achiral or achiral/chiral hyphenated dual column systems.**
- **Selectivity, resolution and retention for the corresponding targets on the dual column systems are summarized in Tables 2, 3, and 4, respectively.**
- **Of the mixtures tested, all three dual achiral/achiral column coupling systems shown reveal an increased retention but their effect on the selectivity of the analytes are mixed.**
 - **In the case of Mixture 1, target separation was improved only with the combination of 4-EP → Benzamide.**
 - **For Mixture 2, all three achiral/achiral dual column systems provided better separation than 4-EP alone.**
 - **In Mixture 3, target separation in all three dual column systems was compromised with no separation observed for the 4-EP → 2CN/Diol combination ($\alpha = 1.0$).**
- **Although the chiral IA column (100 mm) has a shorter length than the three other achiral columns (150 mm) tested, the dual achiral/chiral combination of 4-EP → IA provided the strongest retention, a significant improvement in selectivity ($\alpha \geq 1.2$) and the best separation of the mixtures.**
 - **Elution order in Mixture 1 by 4-EP → IA was reversed in comparison with 4-EP and three other combinations.**
 - **Similar column retention and improved chromatographic separation have also been observed in our use of chiral stationary phases for SFC purification of complex mixtures which show little or no separation in reversed phase HPLC and on achiral SFC stationary phases.[2,3]**

Fig 2. SFC-MS Analytical Profile of Mixture 1

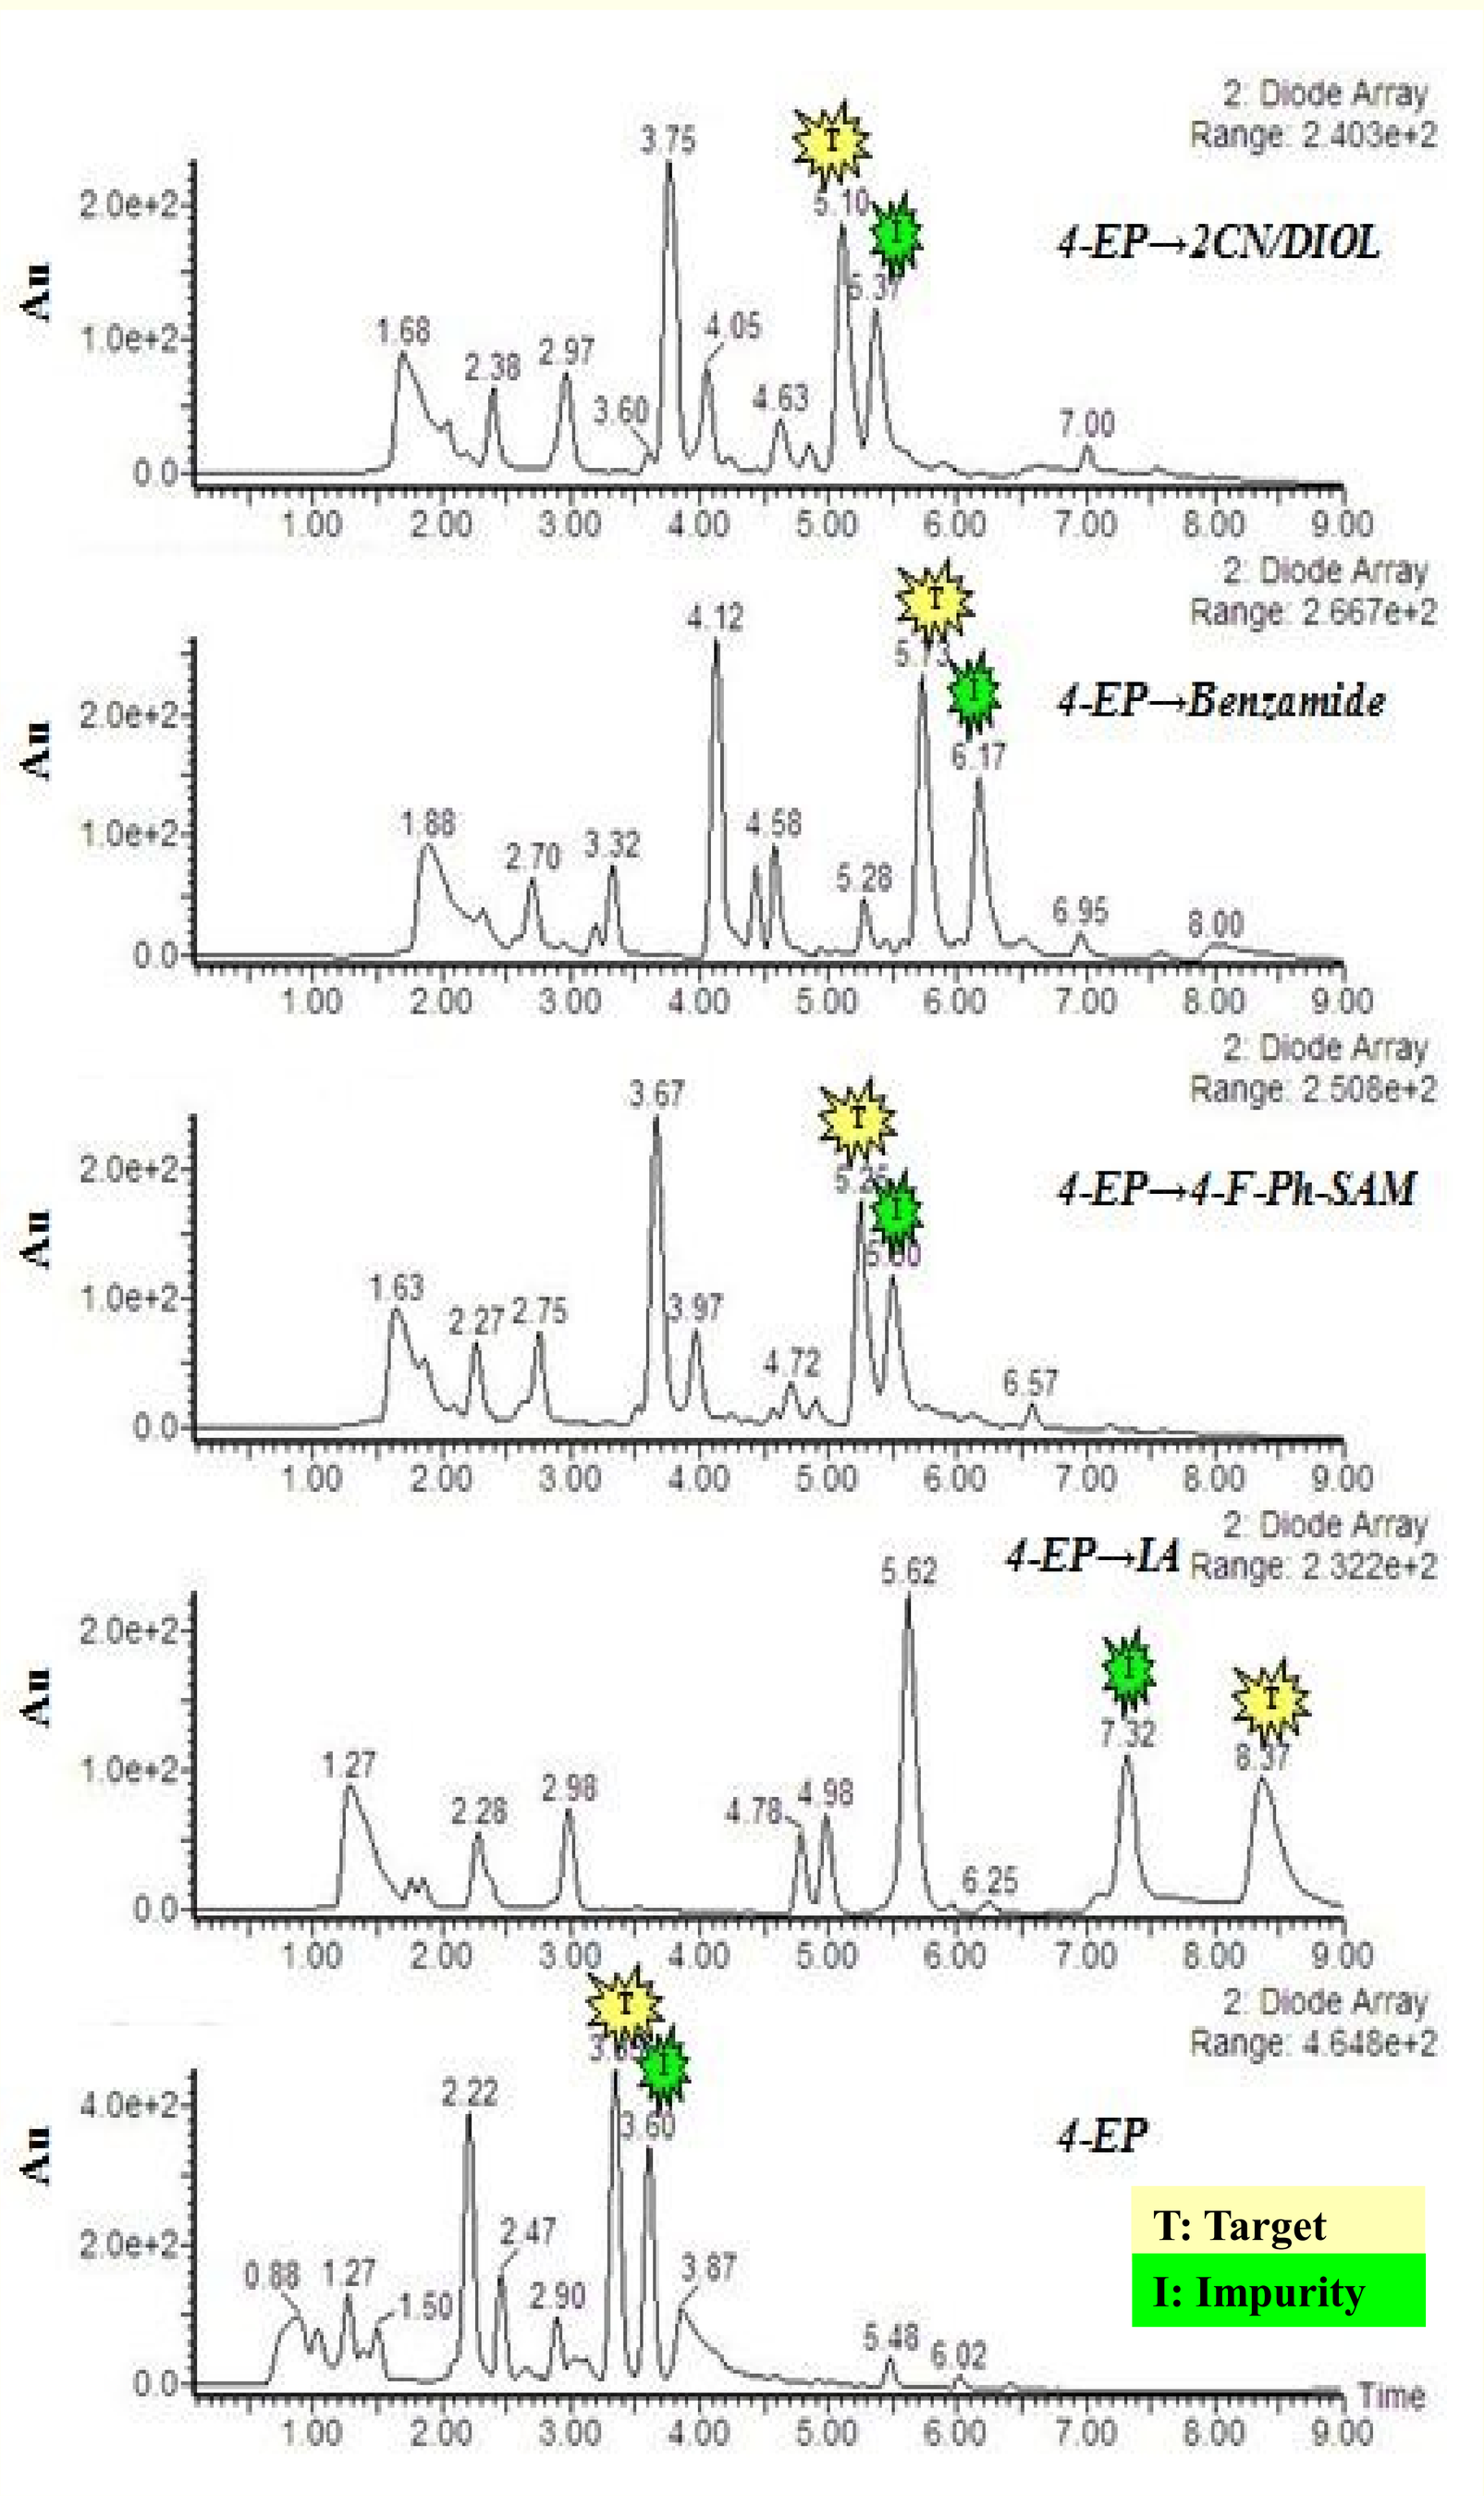
4-EP→2CN/DIOL
4-EP→Benzamide
4-EP→4-F-Ph-SAM
4-EP→IA
4-EP
T: Target
I: Impurity
2: Diode Array
Time
Au

Fig 3. SFC-MS Analytical Profile of Mixture 2

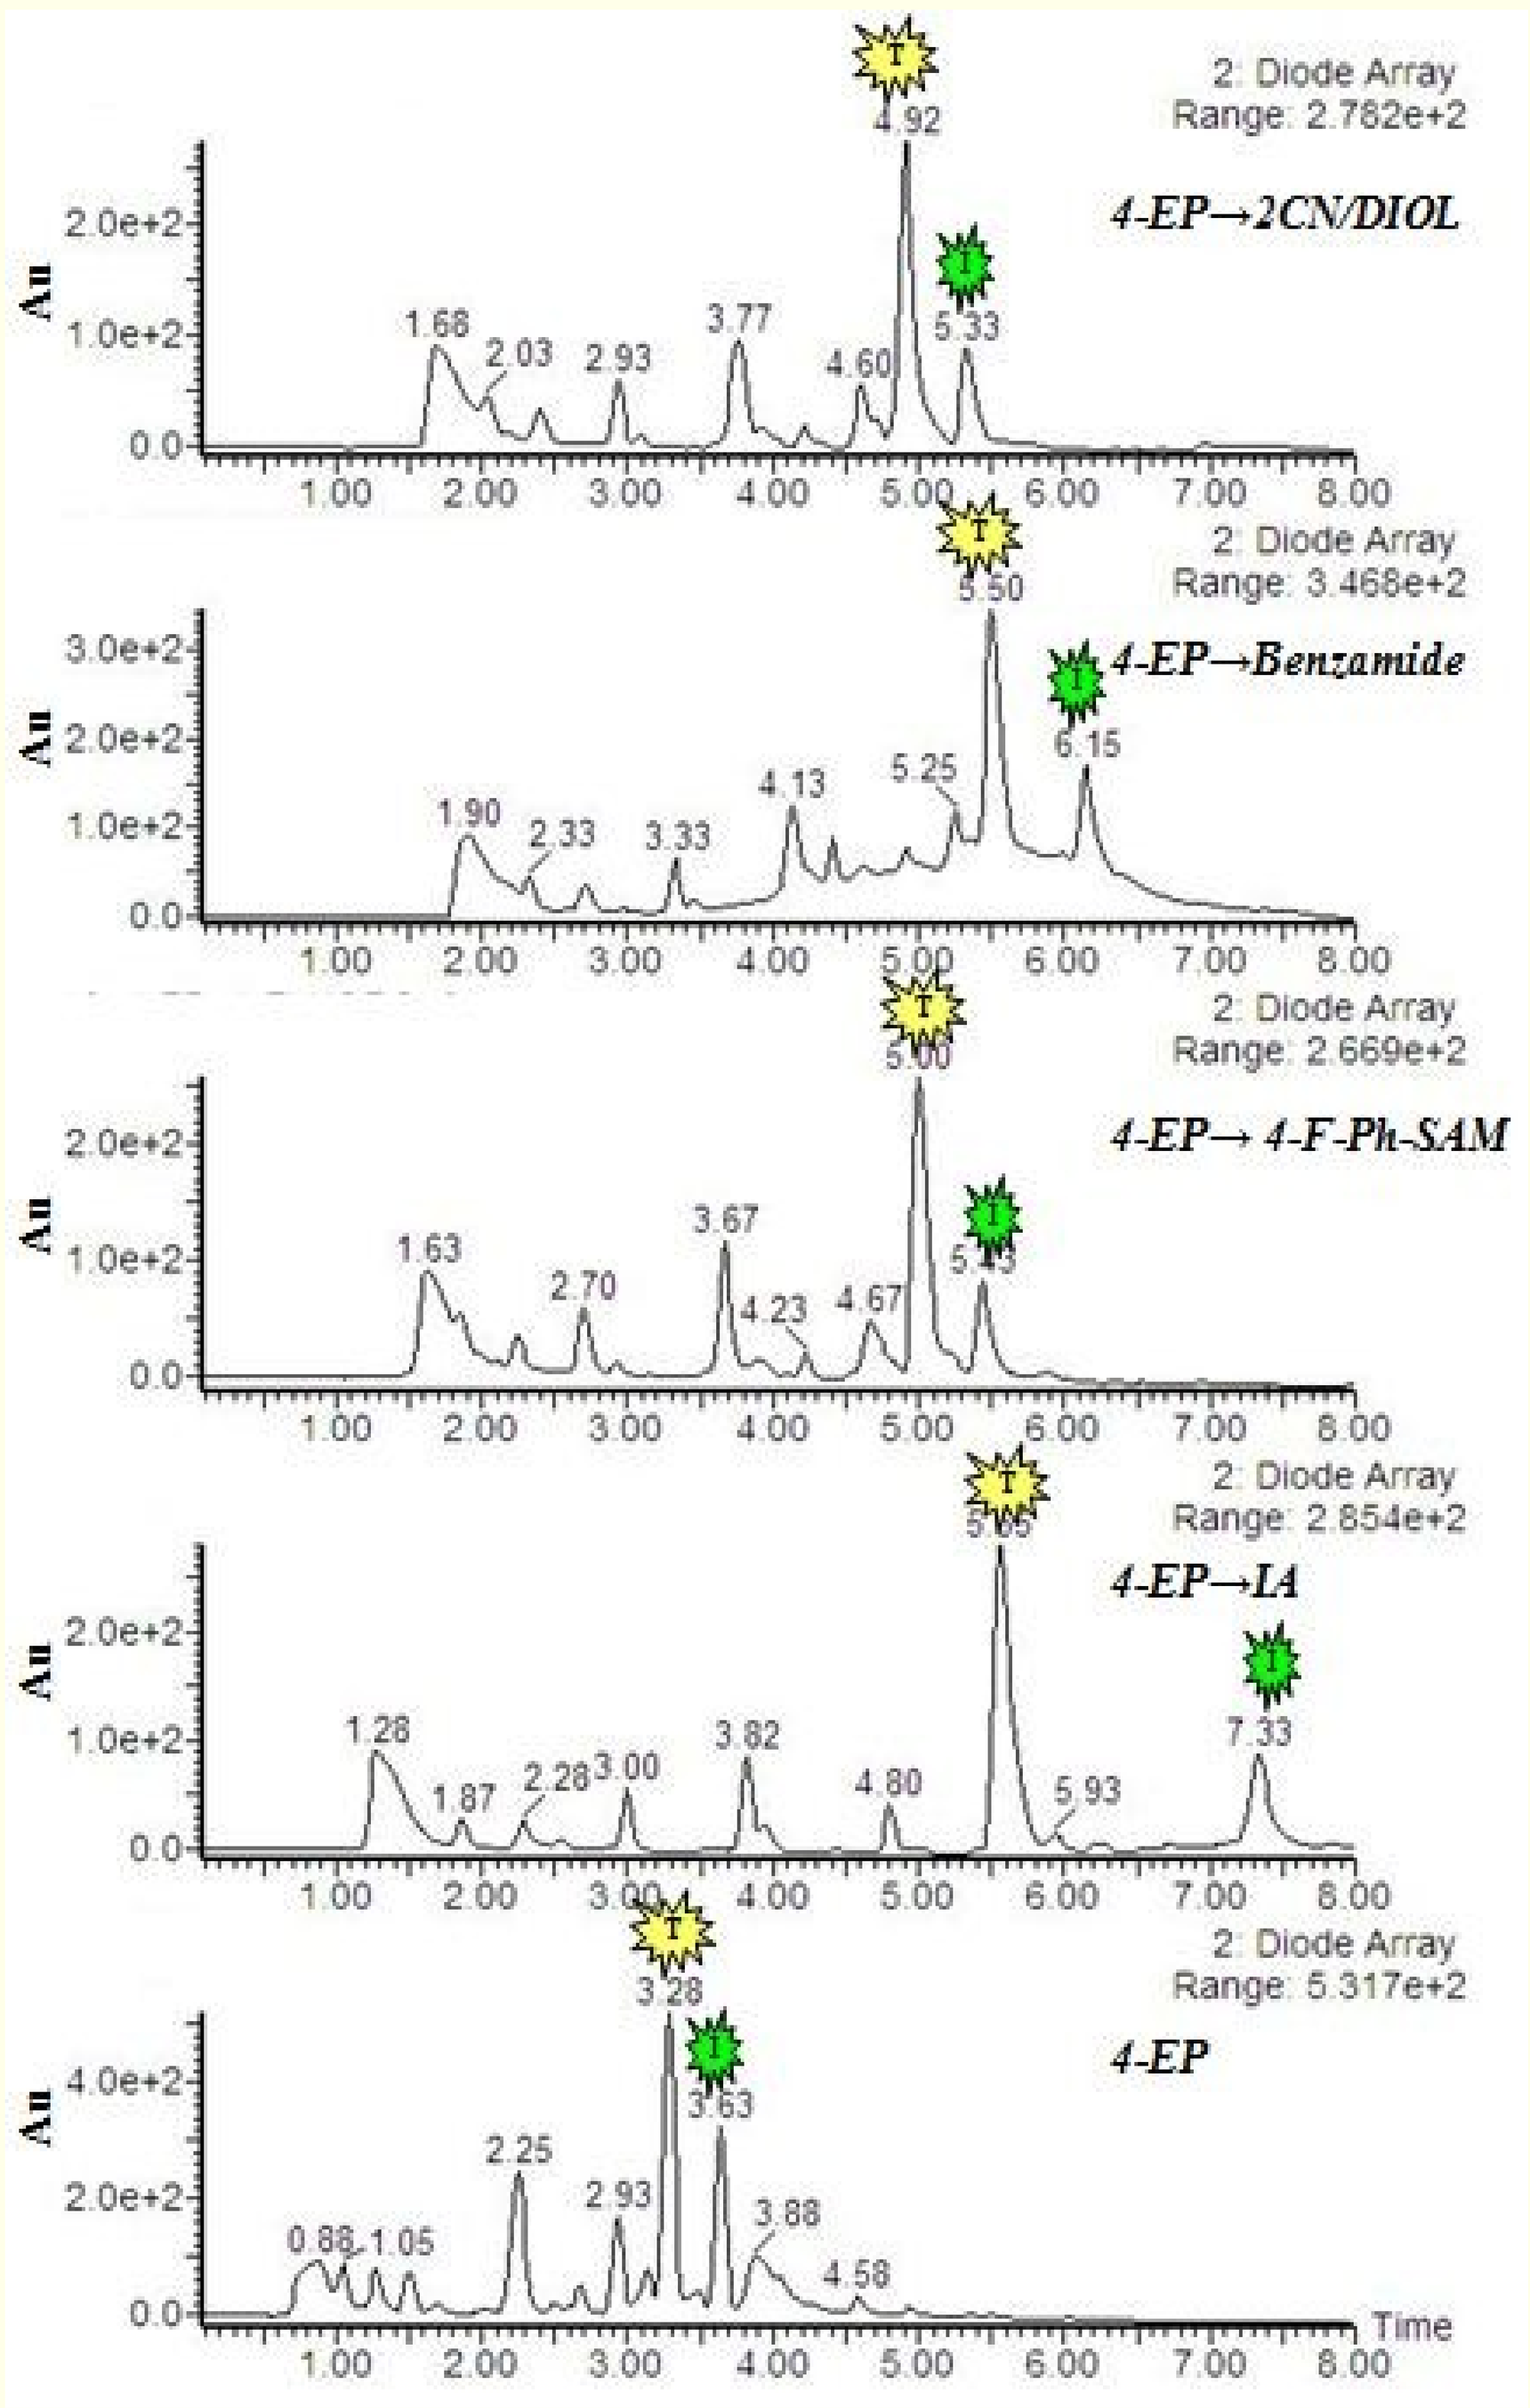
4-EP→2CN/DIOL
4-EP→Benzamide
4-EP→ 4-F-Ph-SAM
4-EP→IA
4-EP
2: Diode Array
Time
Au

Fig 4. SFC-MS Analytical Profile of Mixture 3

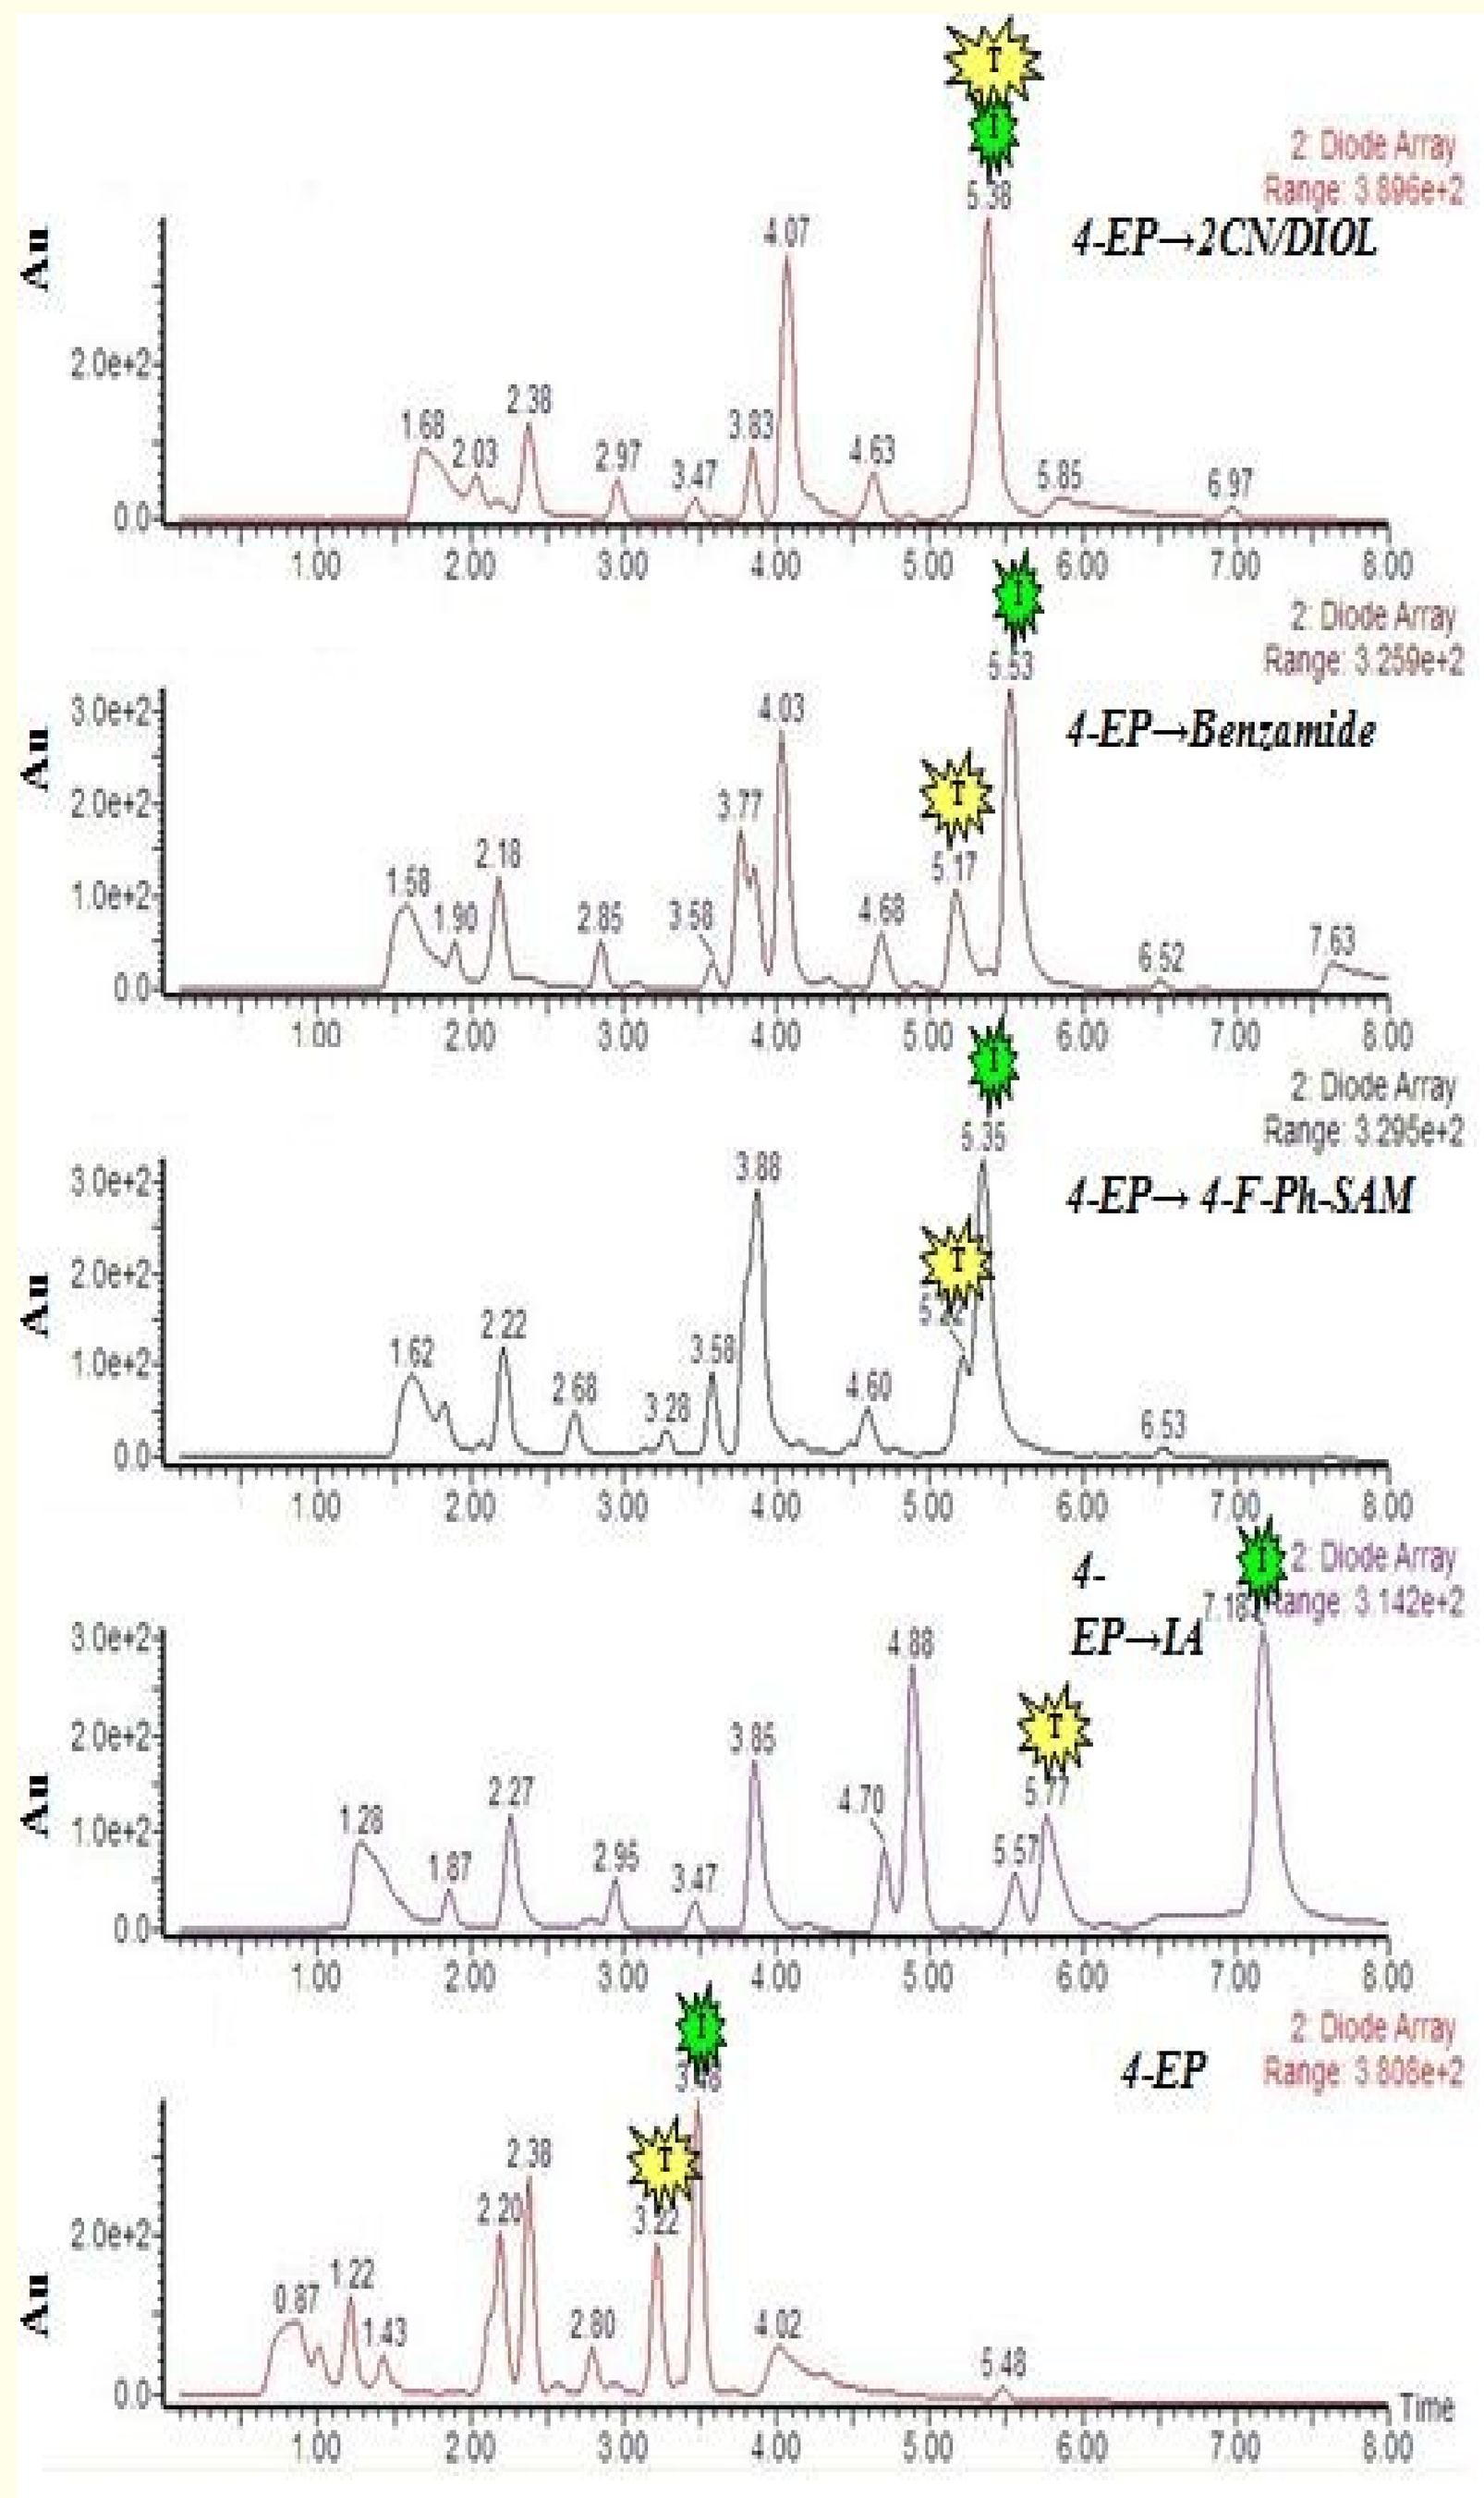
4-EP→2CN/DIOL
4-EP→Benzamide
4-EP→ 4-F-Ph-SAM
4-EP→IA
4-EP
2: Diode Array
Time
Au

Table 2. Selectivity & Resolution Data of Mixture 1

	4-EP	4-EP → Benzamide	4-EP → 4-F-Ph-SAM	4-EP → 2-CN/Diol	4-EP → IA
RT_{Target} (T)	3.35	5.73	5.25	5.10	8.37
$RT_{Impurity}$ (I)	3.60	6.17	5.50	5.37	7.32
α	1.09	1.11	1.07	1.08	1.20
Rs	1.84	2.57	1.01	1.46	3.02

Table 3. Selectivity & Resolution Data of Mixture 2

	4-EP	4-EP → Benzamide	4-EP → 4-F-Ph-SAM	4-EP → 2-CN/Diol	4-EP → IA
RT_{Target} (T)	3.28	5.50	5.00	4.92	5.55
$RT_{Impurity}$ (I)	3.63	6.15	5.43	5.33	7.33
α	1.14	1.18	1.13	1.12	1.42
Rs	1.31	3.07	2.31	2.32	8.05

Table 4. Selectivity & Resolution Data of Mixture 3

	4-EP	4-EP → Benzamide	4-EP → 4-F-Ph-SAM	4-EP → 2-CN/Diol	4-EP → IA
RT_{Target} (T)	3.22	5.17	5.22	5.38	5.77
$RT_{Impurity}$ (I)	3.48	5.53	5.35	5.38	7.18
α	1.11	1.10	1.00	1.0	1.32
Rs	3.98	1.84	0.59*	0	5.64

*: Partially overlapped

Conclusions

- ❖ **A set of tandem dual stationary phases was investigated and their chromatographic performances were evaluated.**
- ❖ **The 4-EP → Benzamide combination performed better or similar to 4-EP alone and is a suitable dual achiral/achiral combination in terms of separation power and resolution.**
 - **Other dual achiral/achiral column combinations tend to increase column retention without improving separation.**
 - **Our preliminary data implies that the separation of compound libraries depends on multiple parameters, including the prerequisite for a high degree of selectivity which is derived from the "primary column" (4-EP) and its synergy with the "orthogonal" column.**
- ❖ **The 4-EP → IA dual achiral/chiral stationary phase system was superior in column retention and improved chromatographic efficiency.**
 - **Some of the "un-resolvable mixtures" were successfully separated.**
 - **Overall separation performances make it an effective column system for the semi-preparative purification of larger libraries (ongoing).**
 - **The benefits greatly offset the longer chromatographic time and higher costs associated with the chiral stationary phase.**
- ❖ **Improved separation in many column combinations has also justified further studies to include a more diverse set of achiral and chiral stationary phases and their combination with 4-EP for achiral SFC purification of compound libraries.**

References

1. L. Ma, V. Lazarescu and M. J. Mulvihill: "Toward a Universal Approach for Mass-Directed SFC Purification of Small Molecule Compound Libraries", presented to the 4th International Conference on Packed-Column SFC in Stockholm, Sweden, September 15-16, 2010.
2. V. Lazarescu, M. J. Mulvihill and L. Ma: "Case Studies on Compound-Specific Achiral SFC Purification", poster presented to the 4th International Conference on Packed-Column SFC in Stockholm, Sweden, September 15-16, 2010.
3. V. Lazarescu, M. J. Mulvihill and L. Ma: "A Generic Workflow for Achiral SFC Purification of Complex Pharmaceutical Mixtures", LCGC North America, 2011, *29(5)*, 438-444.
4. F. Mannerino, Z. Ali and J. Wheeler : "Coupling Chiral and Achiral Columns in Series for a Multi-Phase Approach for the Separation of Stereoisomers using Supercritical Fluid Chromatography", poster presented to the 4th International Conference on Packed-Column SFC in Stockholm, Sweden, September 15-16, 2010.

(OSI) oncology

A wholly owned subsidiary of Astellas Pharma, Inc.